Development of an Australian-sourced bilayer collagen membrane for dental guided bone regeneration

Brent Allan[1], Tao Wang[2], Euphemie Bassonga[2], Clair Lee[3], Ming Hao Zheng[2]

[1]Oral Maxillo-Facial Surgery, St John of God Subiaco Hospital, WA, Australia
[2]Centre for Orthopaedic Translational Research, School of Biomedical Science, The University of Western Australia, WA, Australia
[3]Orthocell Ltd, WA, Australia

The University of Western Australia
Achieve International Excellence

Background & Aim

- **Severe osseous defects are often caused by tooth extraction or loss, resulting in the deterioration of the original ridge dimension**
- **Preservation and restoration of bone volume are essential to facilitate subsequent placement of dental implants.**
- **The area of missing bone (defect/void) is often filled with a natural bone mineral (NBM) materials after placement of an appropriately-sized titanium implant**
- **Use of collagen membrane is necessary to direct the regeneration of bone tissue as it prevents the defect being filled by the faster growing gingival tissues.**
- **CelGro™ is a bilayer, acellular type I collagen matrix of porcine origin.**

Methods

- **The native bilayer collagen structure of the porcine source material was preserved during the patented purification process that removed lipids, nucleic acids, and other cellular impurities. Purity was assessed by imaging and chemical analysis.**
- **Patients (N=10) who fulfilled the study eligibility criteria and were enrolled in the study. Study participants received two-stage dental implant treatment with simultaneous GBR using CelGro™ and void-filling material (natural bone mineral).**
- **Implant sites were allowed to heal for 6 months before re-entry surgery. Mucosal tissue conditions and evidence of wound dehiscence or membrane exposure were recorded during the healing period.**
- **Quality of newly formed bone (QT scale) was assessed at the time of re-entry surgery. Vertical (defect height) and horizontal (facial bone wall thickness) dimensions of the implant site were measured immediately after implant placement (baseline) and prior to re-entry by CBCT scan.**

Results

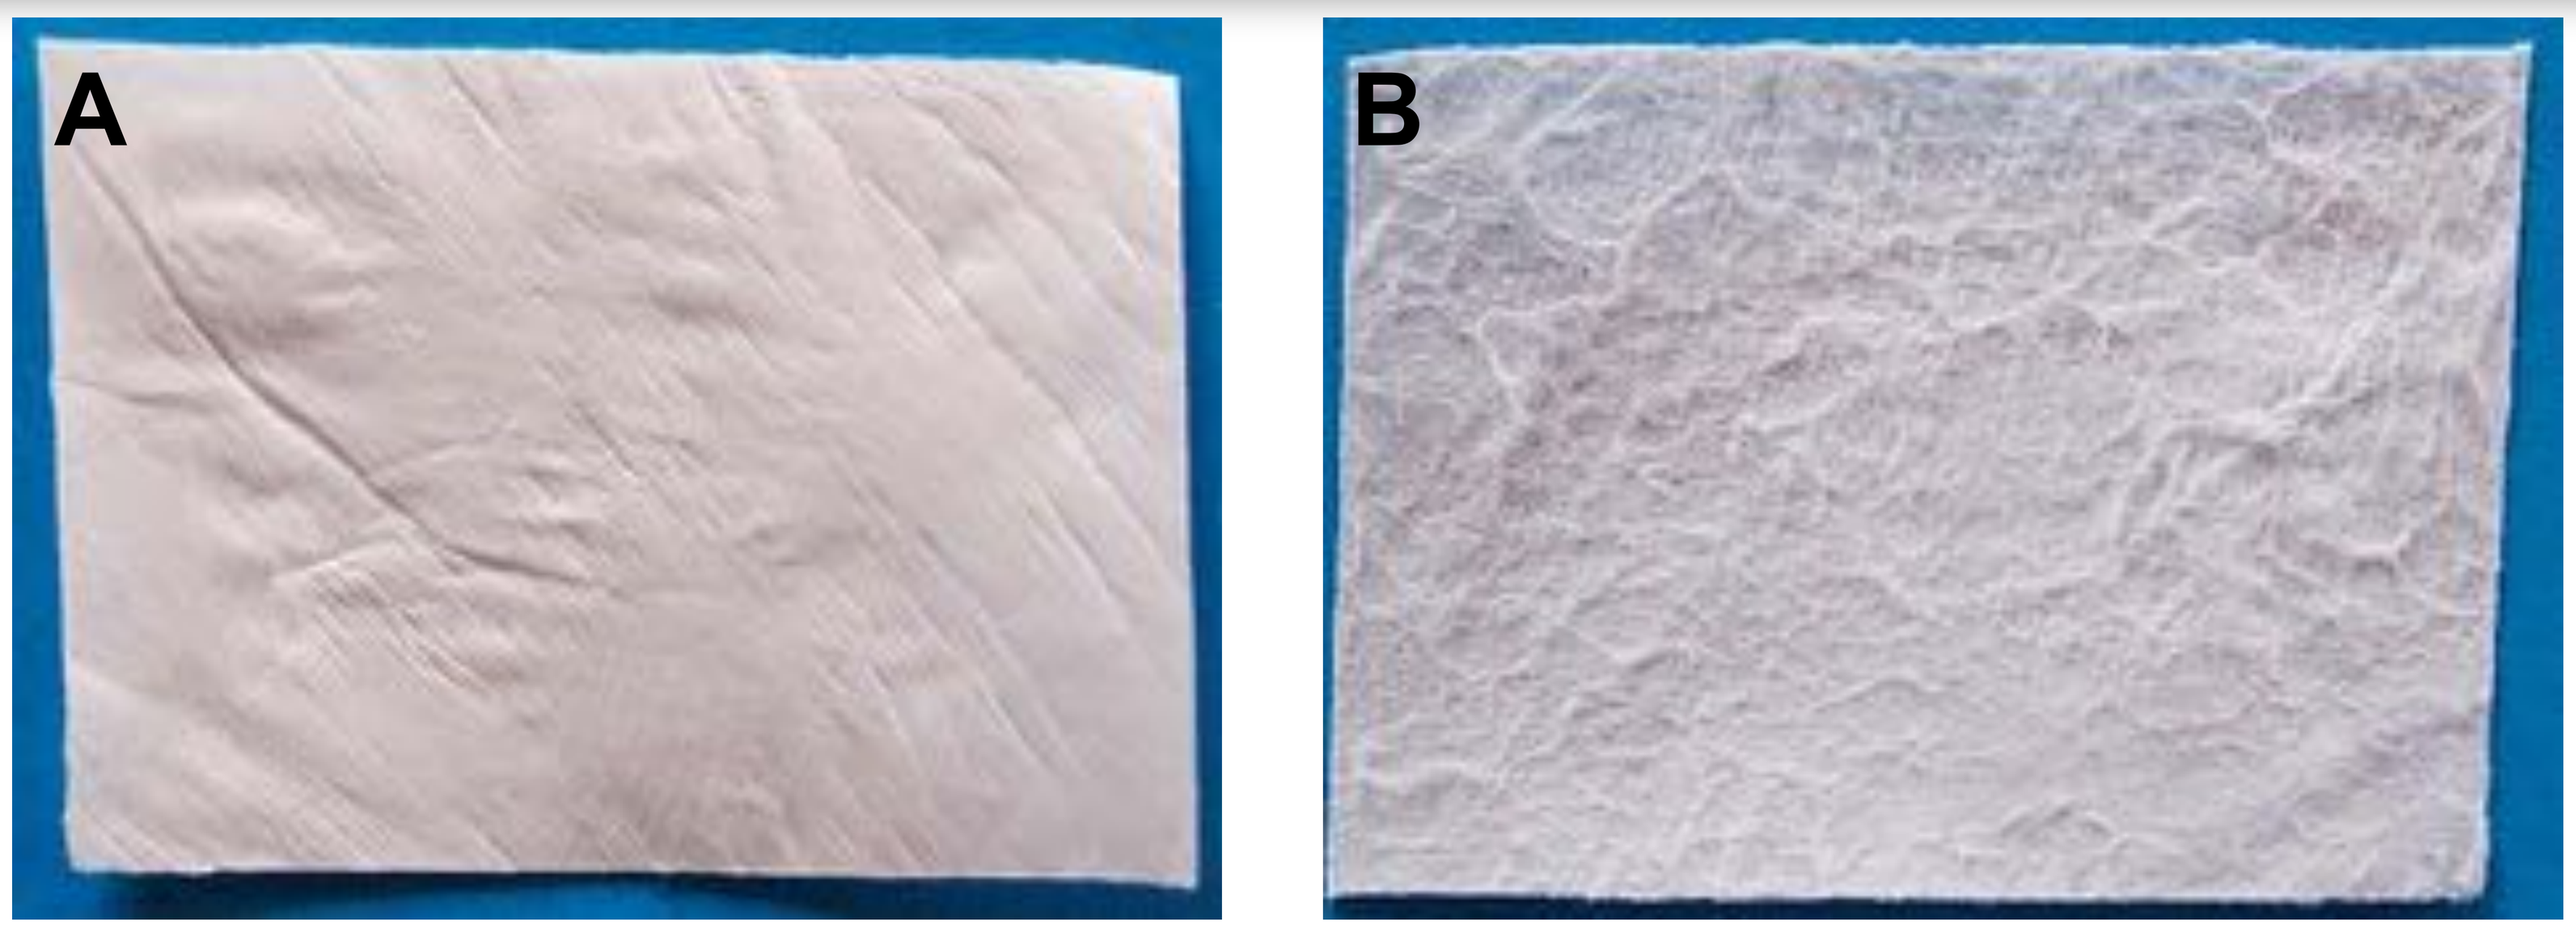
A
B

Fig. 1. Bilayer structure of CelGro™. Visual appearance of the smooth (A) and rough sides (B) of the device.

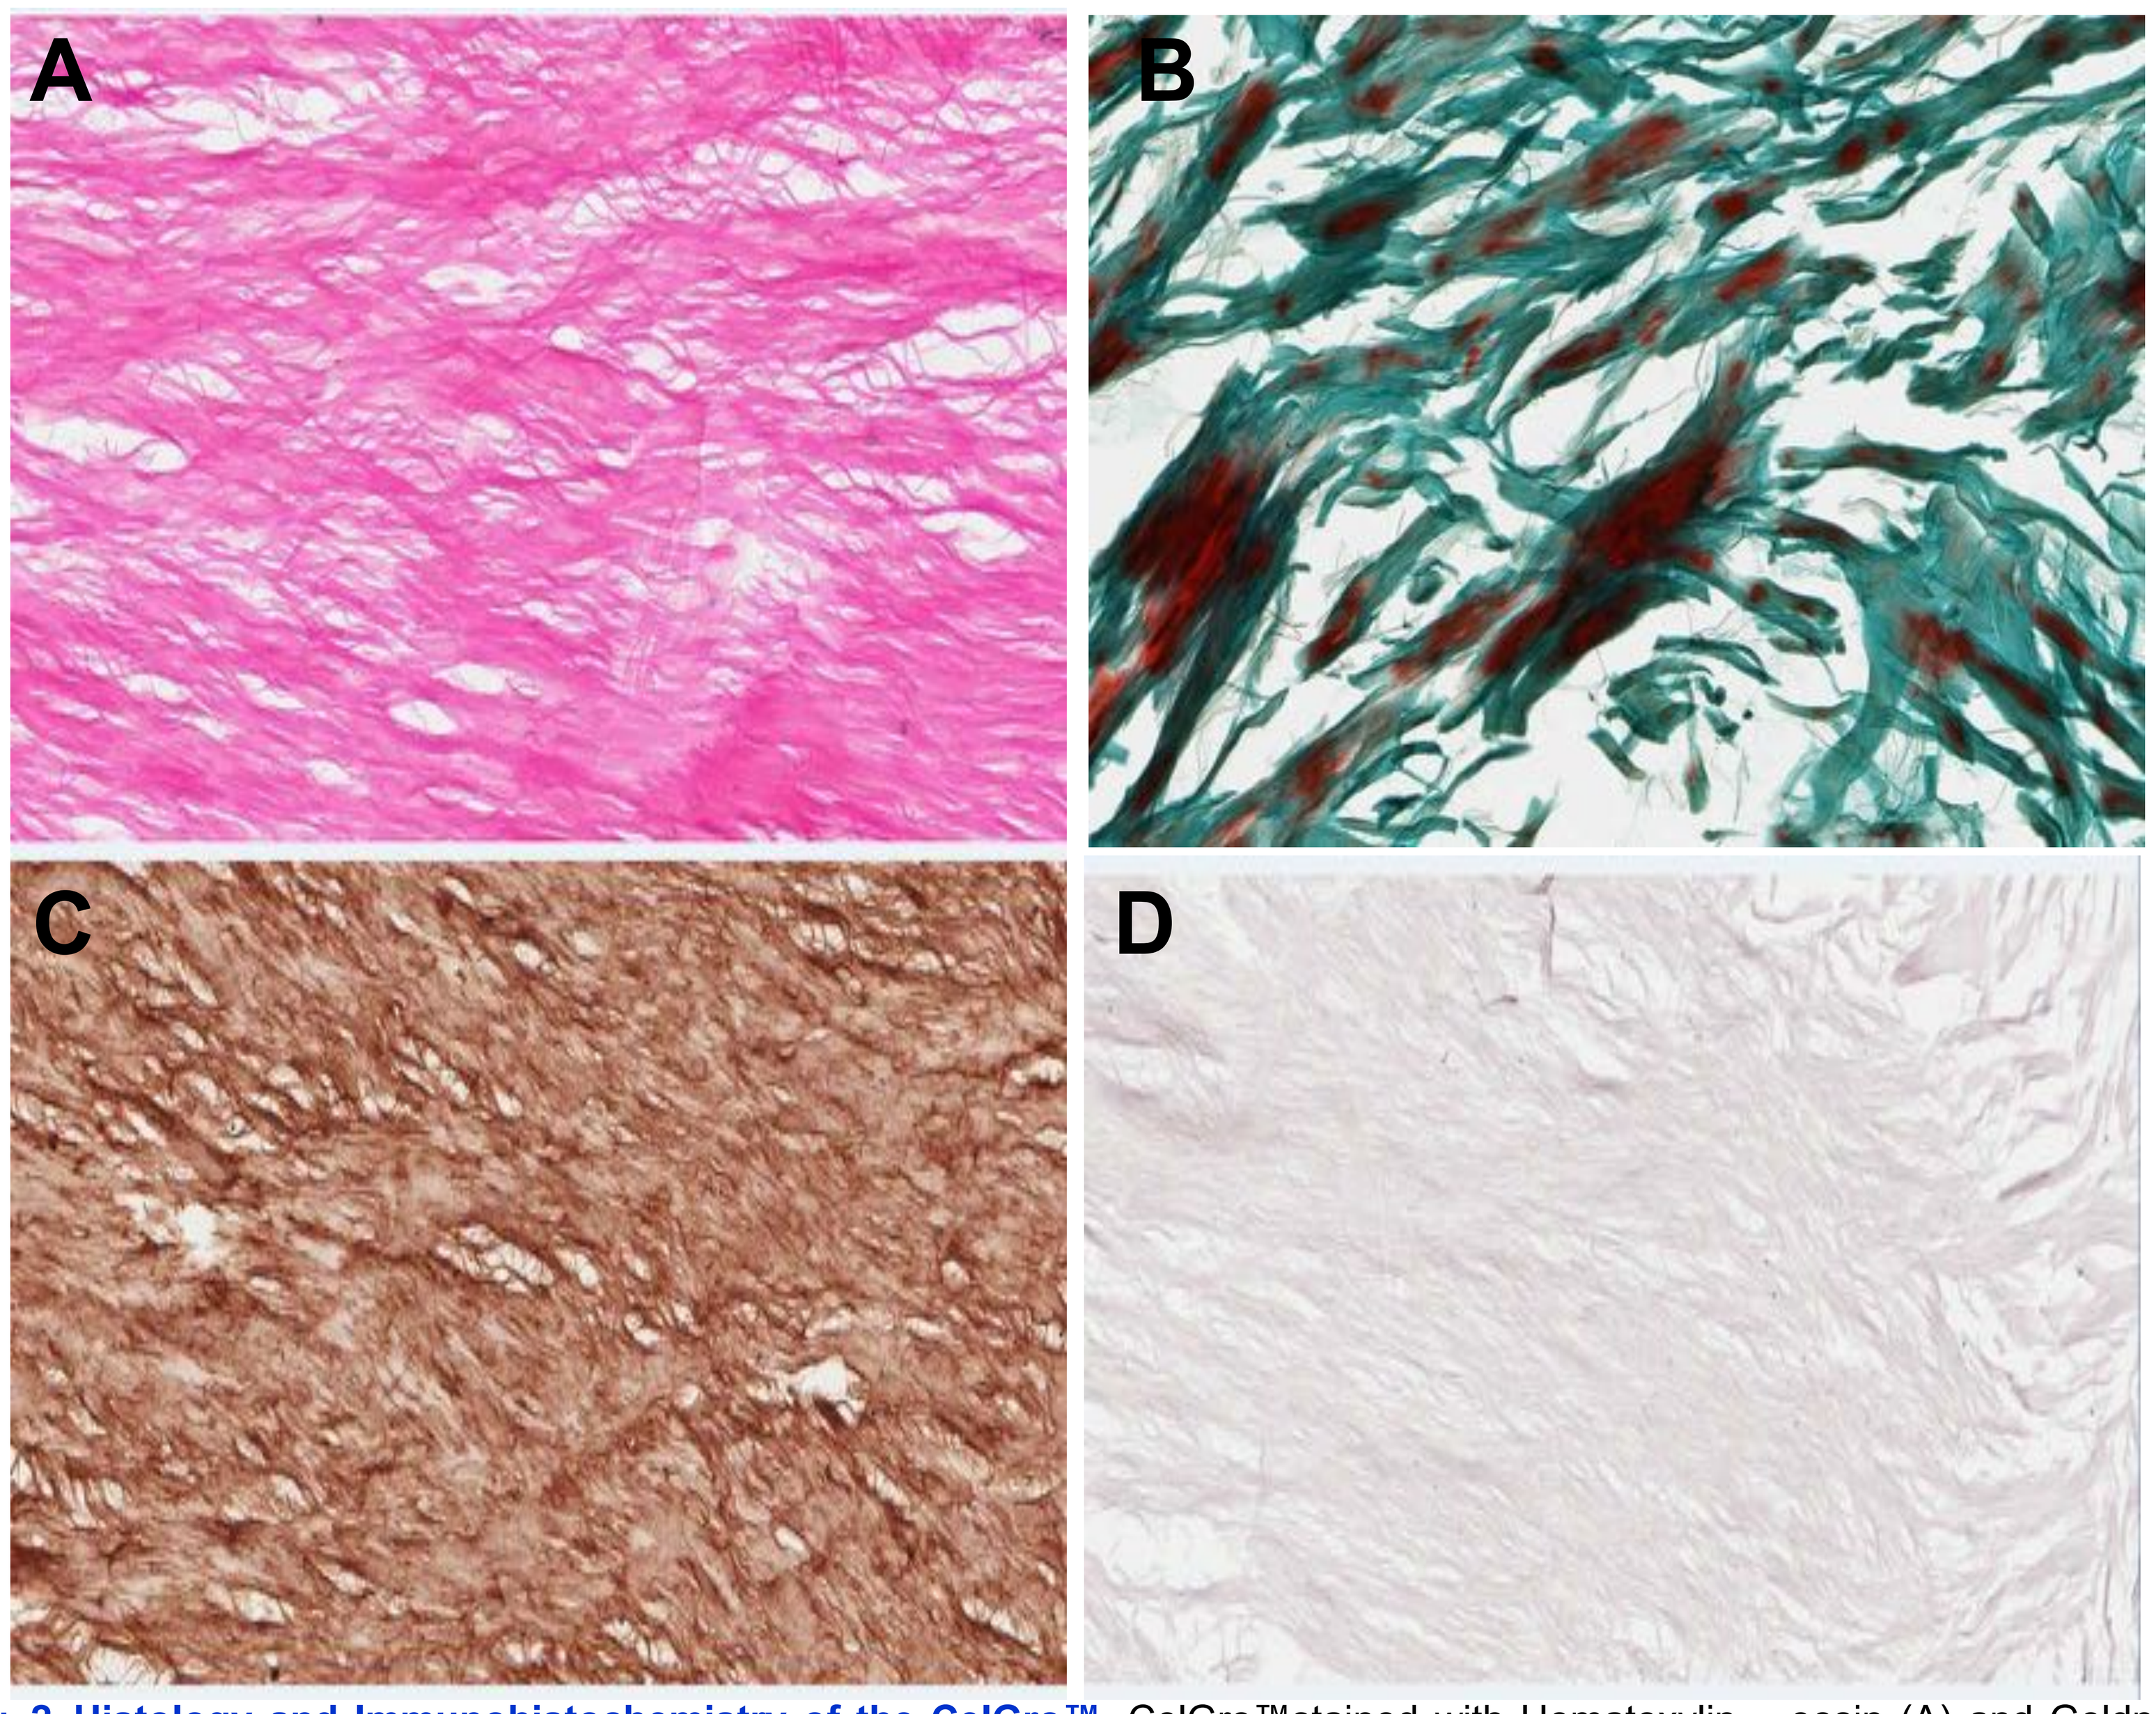
A
B
C
D

Fig. 2. Histology and Immunohistochemistry of the CelGro™. CelGro™stained with Hematoxylin – eosin (A) and Goldner trichrome (B) shows no residue cells. Immunohistochemistry shows a Type I collagen stained brown on CelGro™ (C). No stain was detected for the collagen type III on membrane (D).

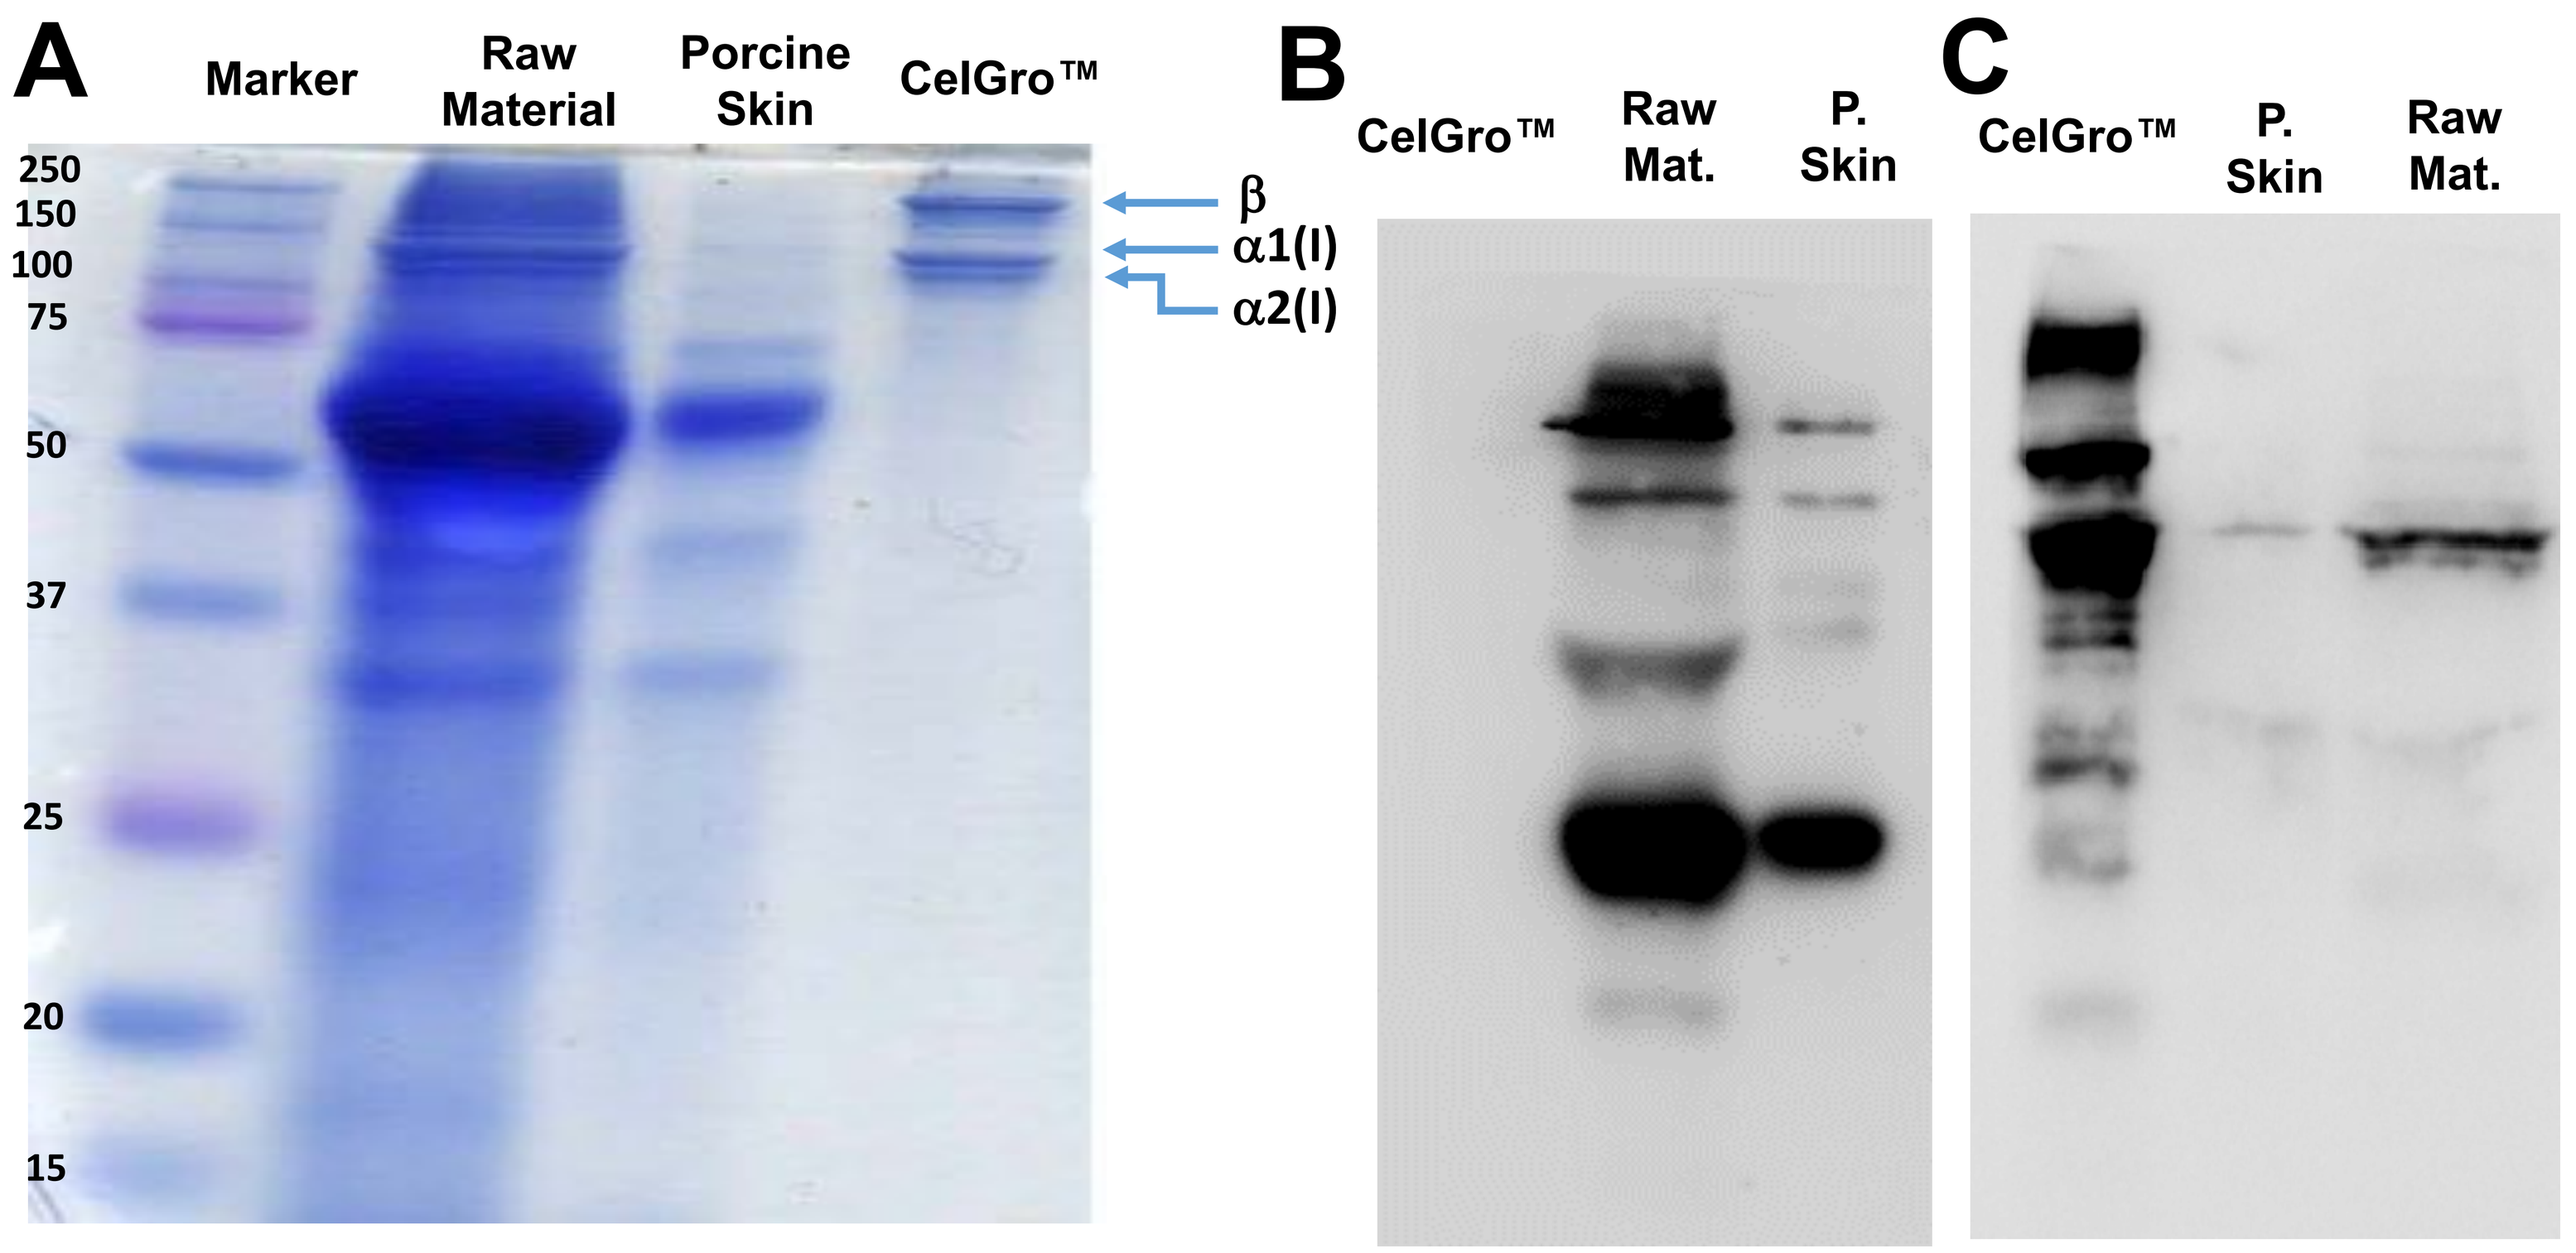
A
Marker
Raw Material
Porcine Skin
CelGro™
250
150
100
75
50
37
25
20
15
β
α1(I)
α2(I)
B
CelGro™
Raw Mat.
P. Skin
C
CelGro™
P. Skin
Raw Mat.

Fig. 3. A. Visualisation of proteins distribution of Raw material, porcine skin and CelGro™ by Coomassie blue stain. The SDS-PAGE gel indicated the presence of bands that match collagens of a low molecular weight between 150 and 100kD (~ 116kDa ; ~ 130 kDa). Electrophorese (SDS-PAGE) of type III Collagen (B) and type I collagen (C) from CelGro™; porcine Raw material; Porcine skin. p

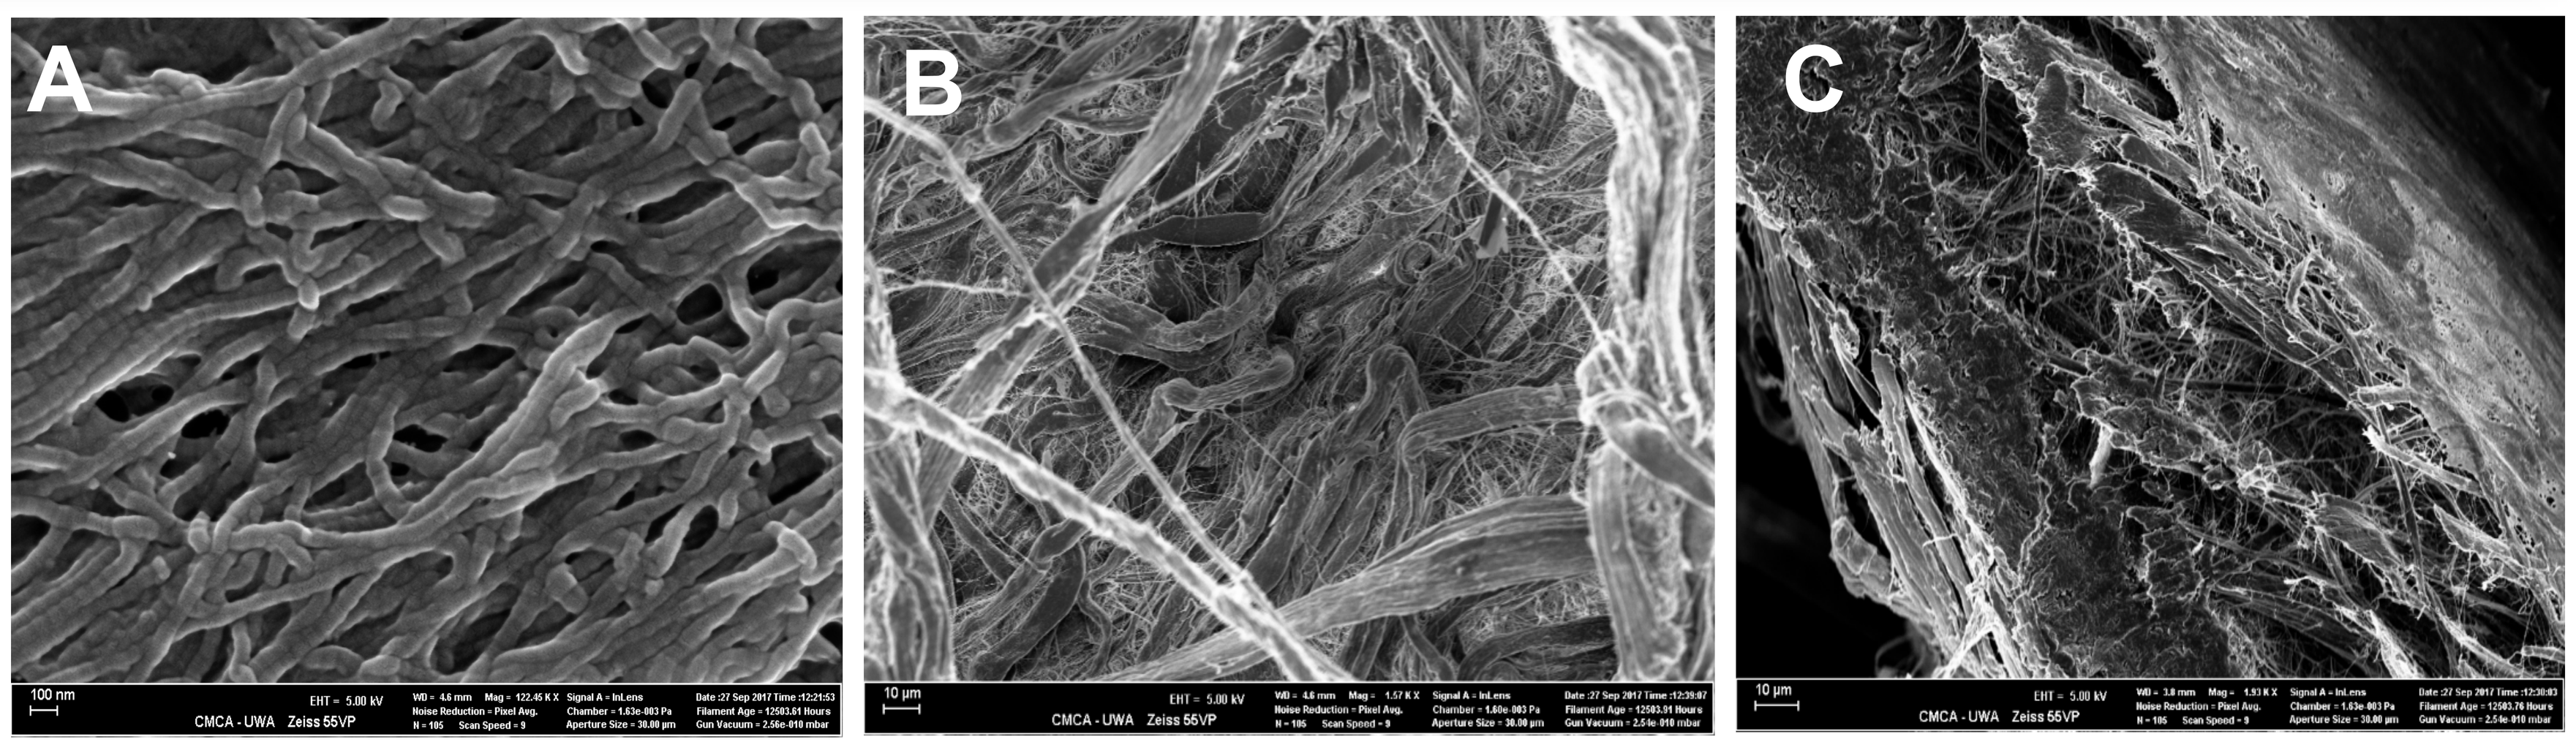
A
B
C
100 nm
10 µm
10 µm

Fig. 4. Collagen fibers structure of CelGro™ by scanning electron microscope (SEM). The aggregation of Type I collagen fibres into bands is well demonstrated on the upper image, smooth surface of the membrane (A). Rough surface of the CelGro collagen membrane (B). Cross section of collagen membrane (C).

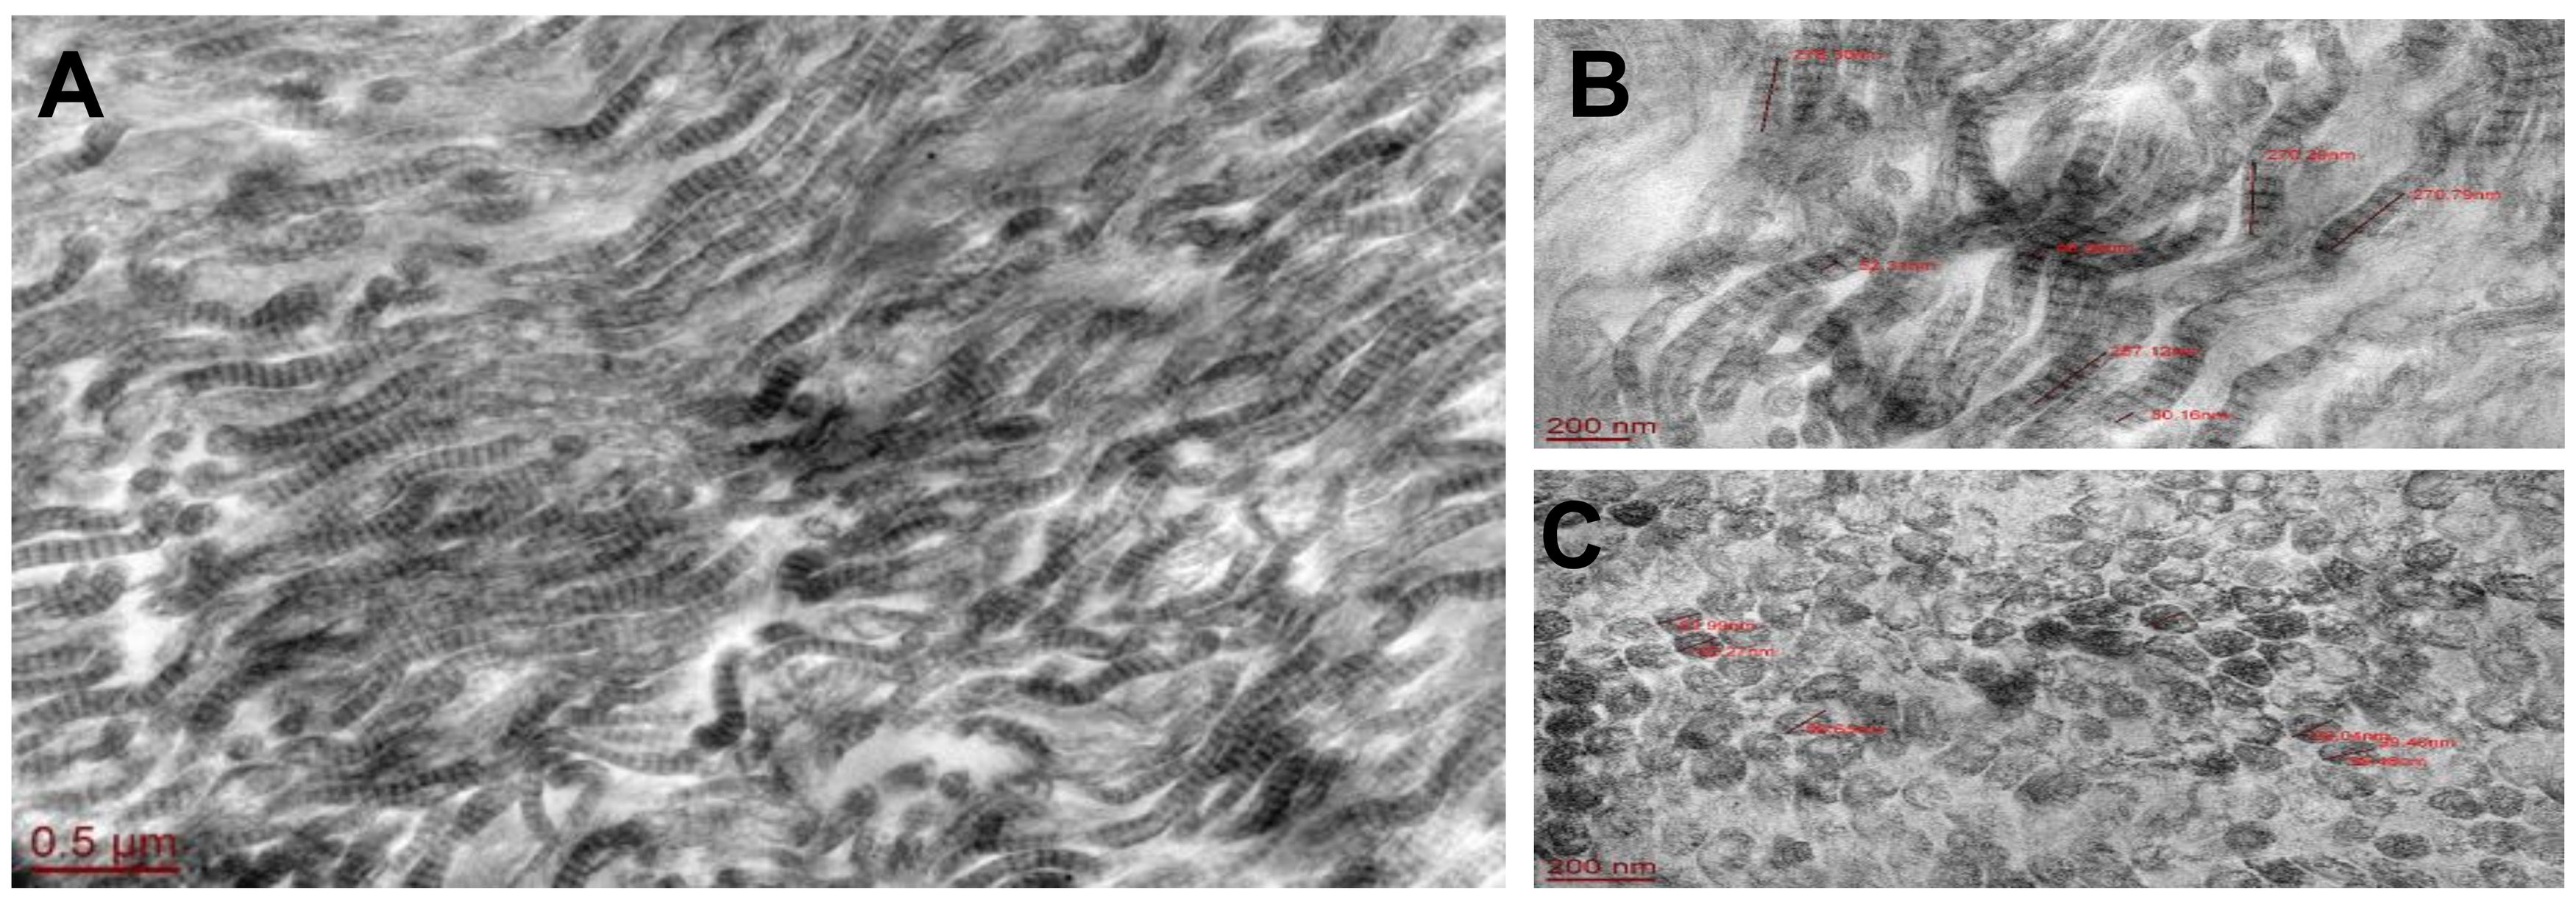
A
B
C
0.5 µm
200 nm
200 nm

Fig. 5. CelGro™ shows the appearance of type I collagen fibres under the transmission electron microscope at different magnifications. Individual fibrils show a characteristic banding pattern which results from the overlap of the tropocollagen constituents. The periodicity of this pattern is typically 53 nm, the Tropocollagen length is measured to be around 300nm. (B). And the diameter of the fibrils ranged from 83-99 nm.

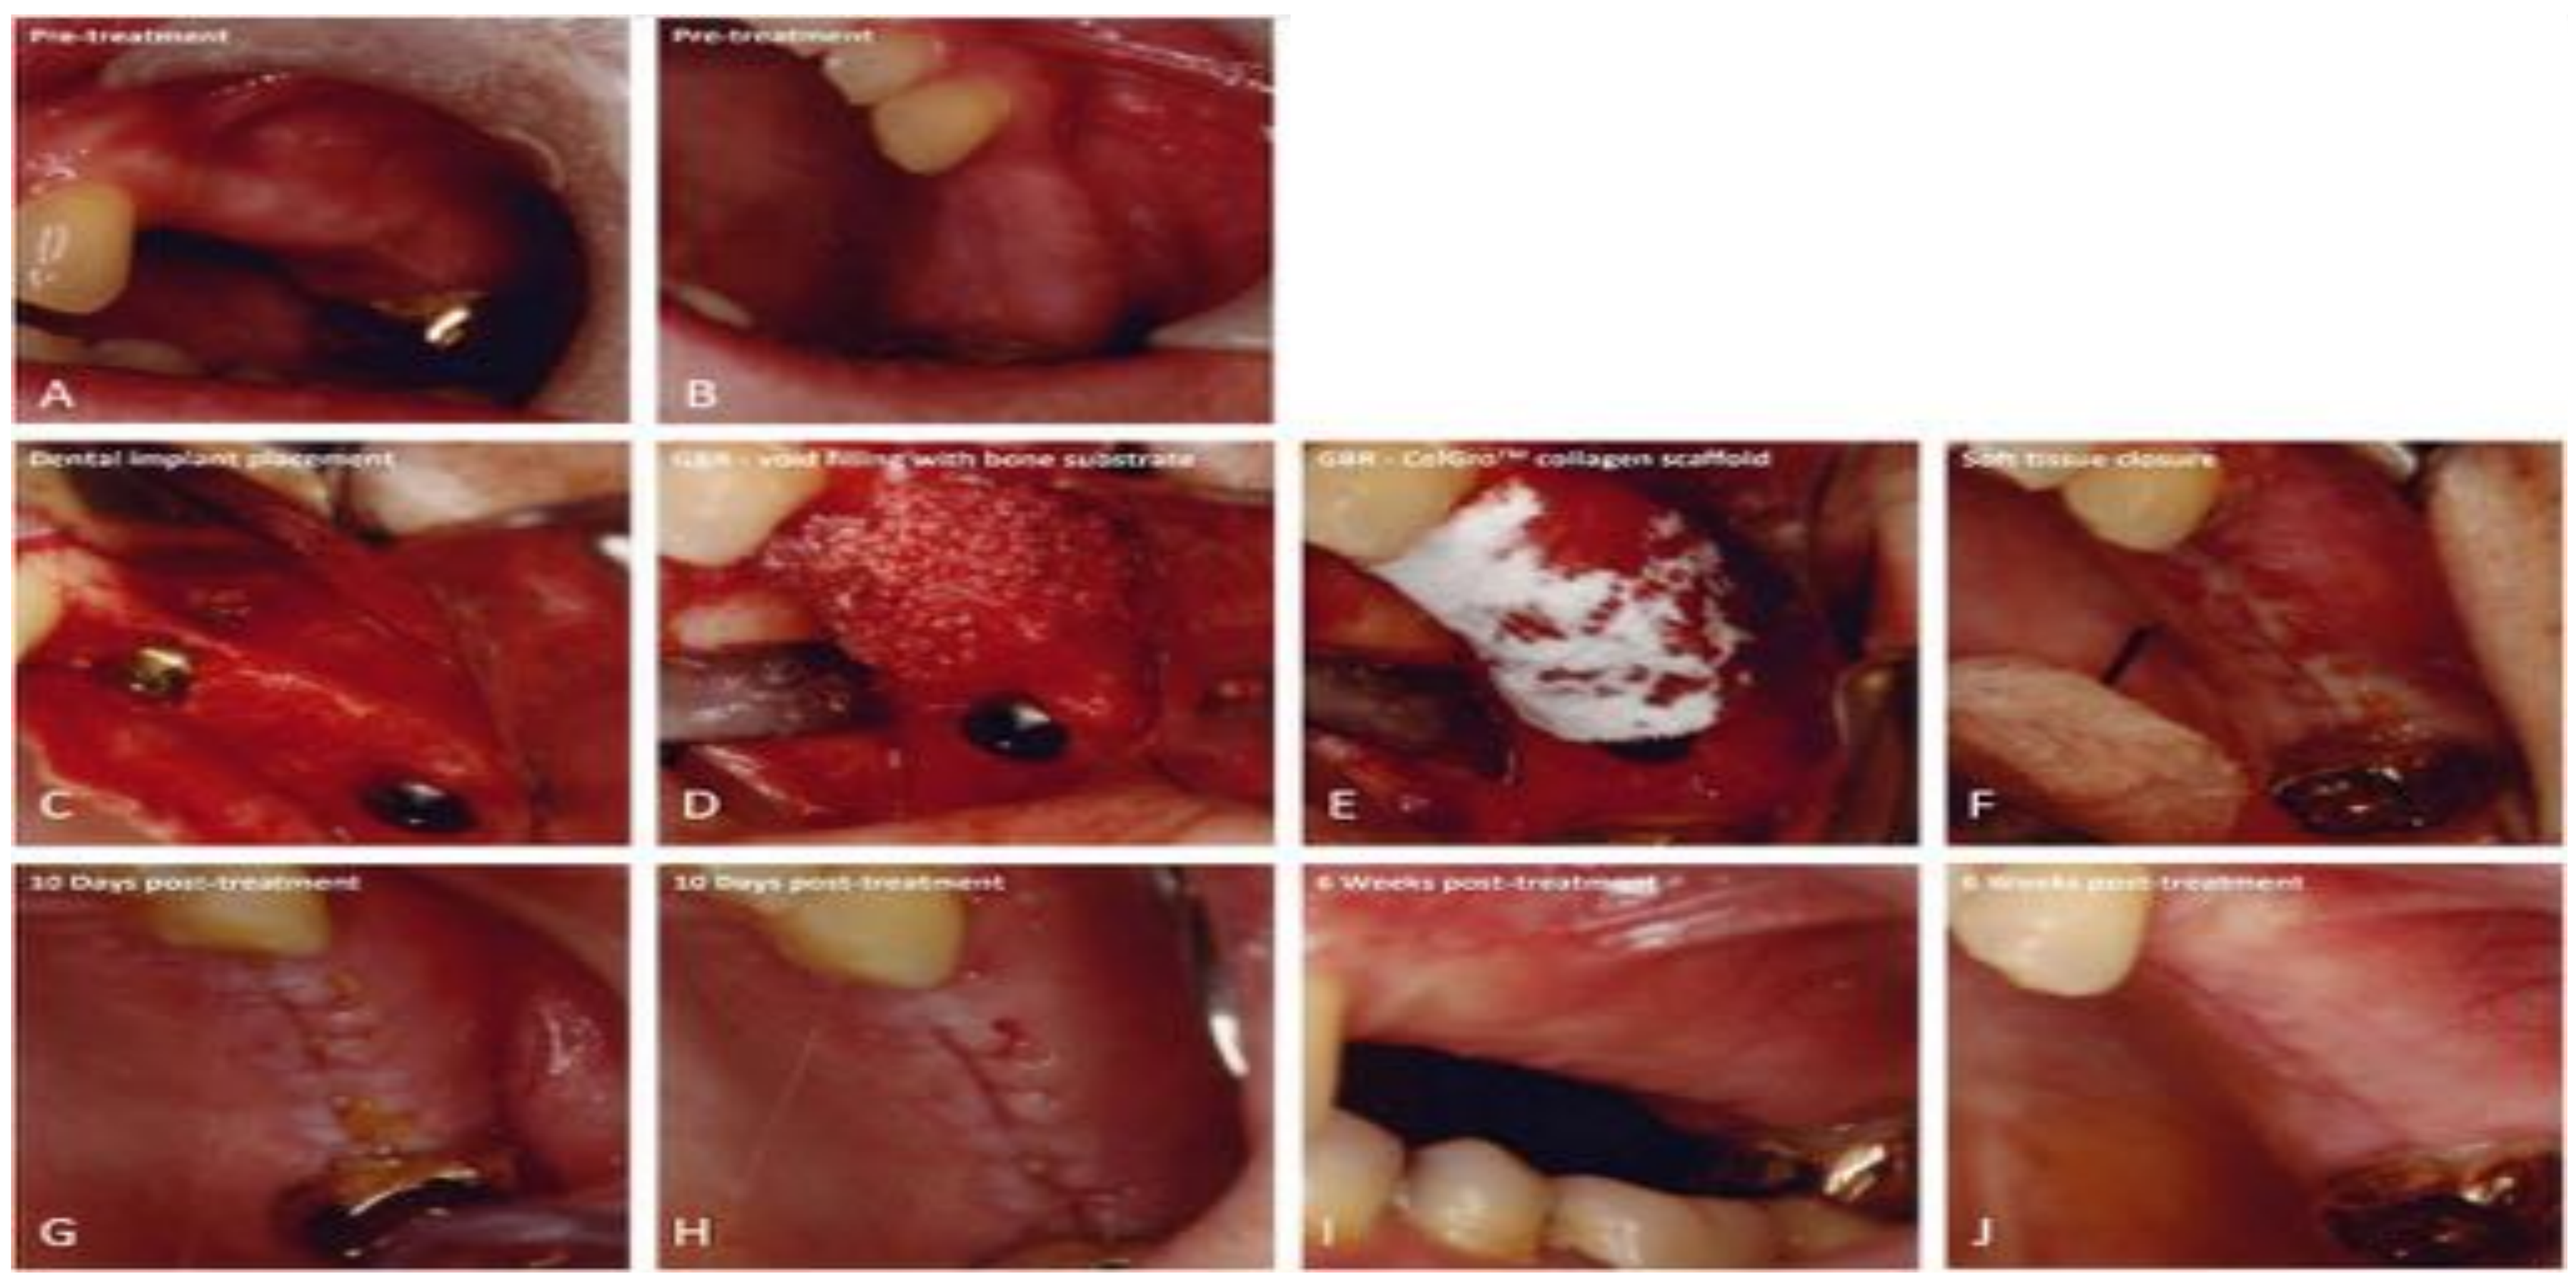
Pre-treatment
Pre-treatment
Dental implant placement
GBR - void filling with bone substrate
GBR - CelGro™ collagen scaffold
Soft tissue closure
10 Days post-treatment
10 Days post-treatment
6 Weeks post-treatment
6 Weeks post-treatment
A
B
C
D
E
F
G
H
I
J

Fig. 6. Representative images of treatment location for participant CG-002-01 before (A,B), during (C-F), 10 days after (G,H) and 6 weeks after (I, J) implant placement

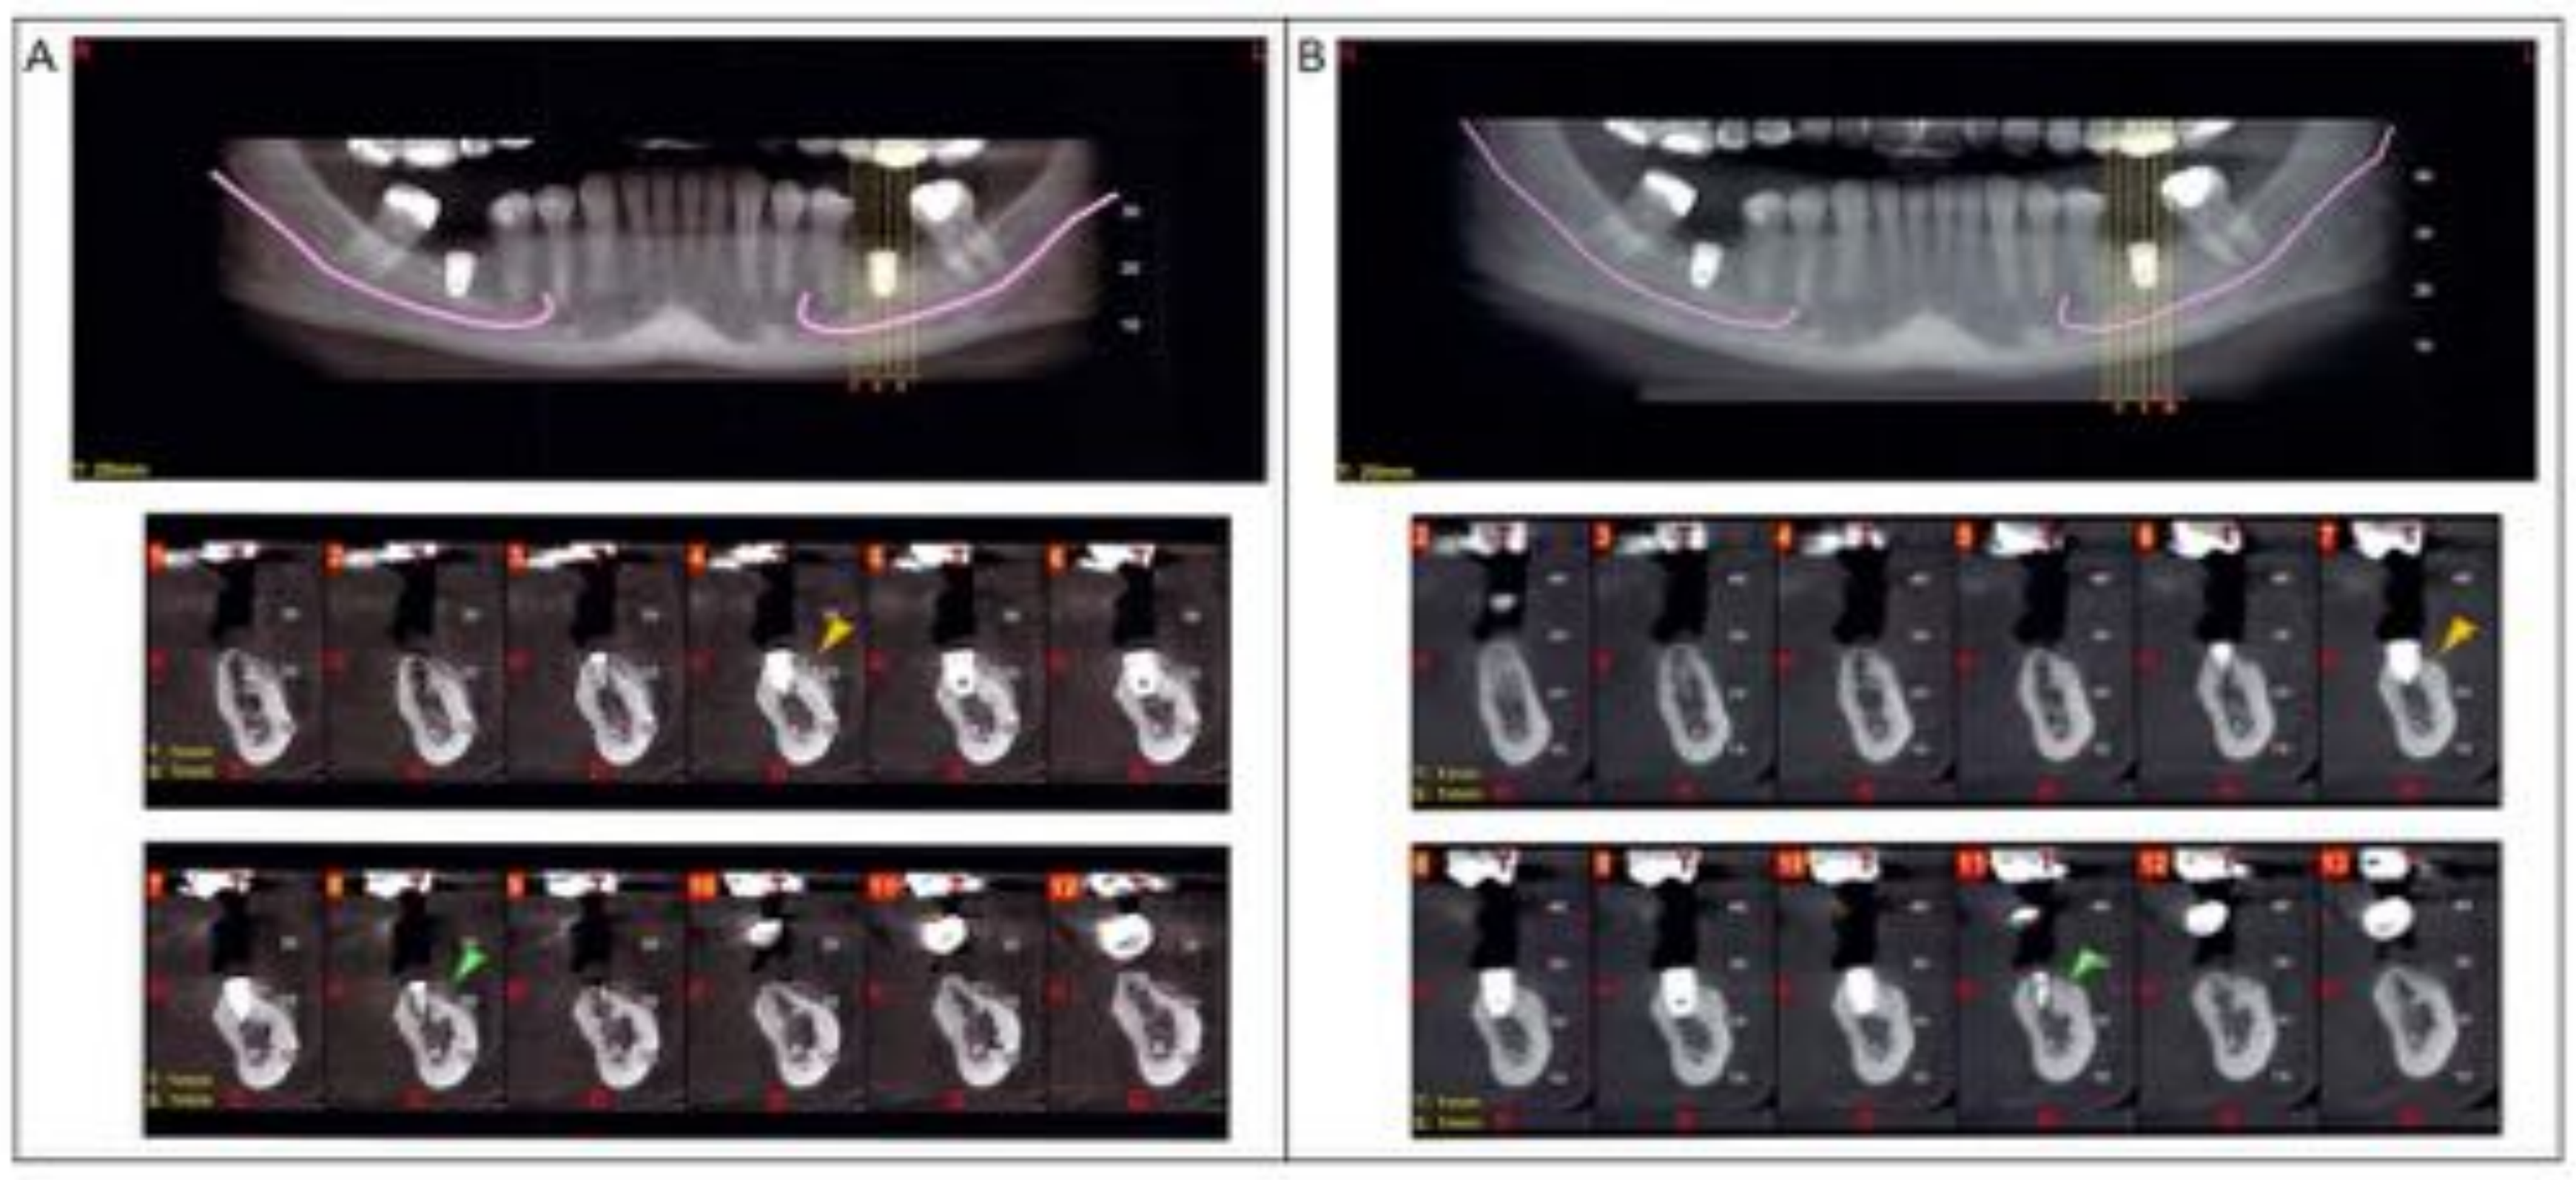
A
B

Fig. 7. Representative Cone Beam Computed Tomography (CBCT) of Patient CG-002-11 showed guided bone regeneration after 6 months. CBCT at post-treatment time-points of 12 days post-implant (A) and 6 months (B). Yellow and green arrowheads represent two identical implant sites in the lower left 2nd premolar and lower left 1st molar regions at 12 days post implant (A) and 6 month (B). Notes that the bone graft material was deposited around the buccal cortex at 12 days post implant (A). New bone formation occurred around the buccal and crestal portion of the implant fixtures in the lower left 2nd premolar and lower left 1st molar regions at 6 month (B). Osseointegration was completed with consolidation of guided bone regeneration in the regions.

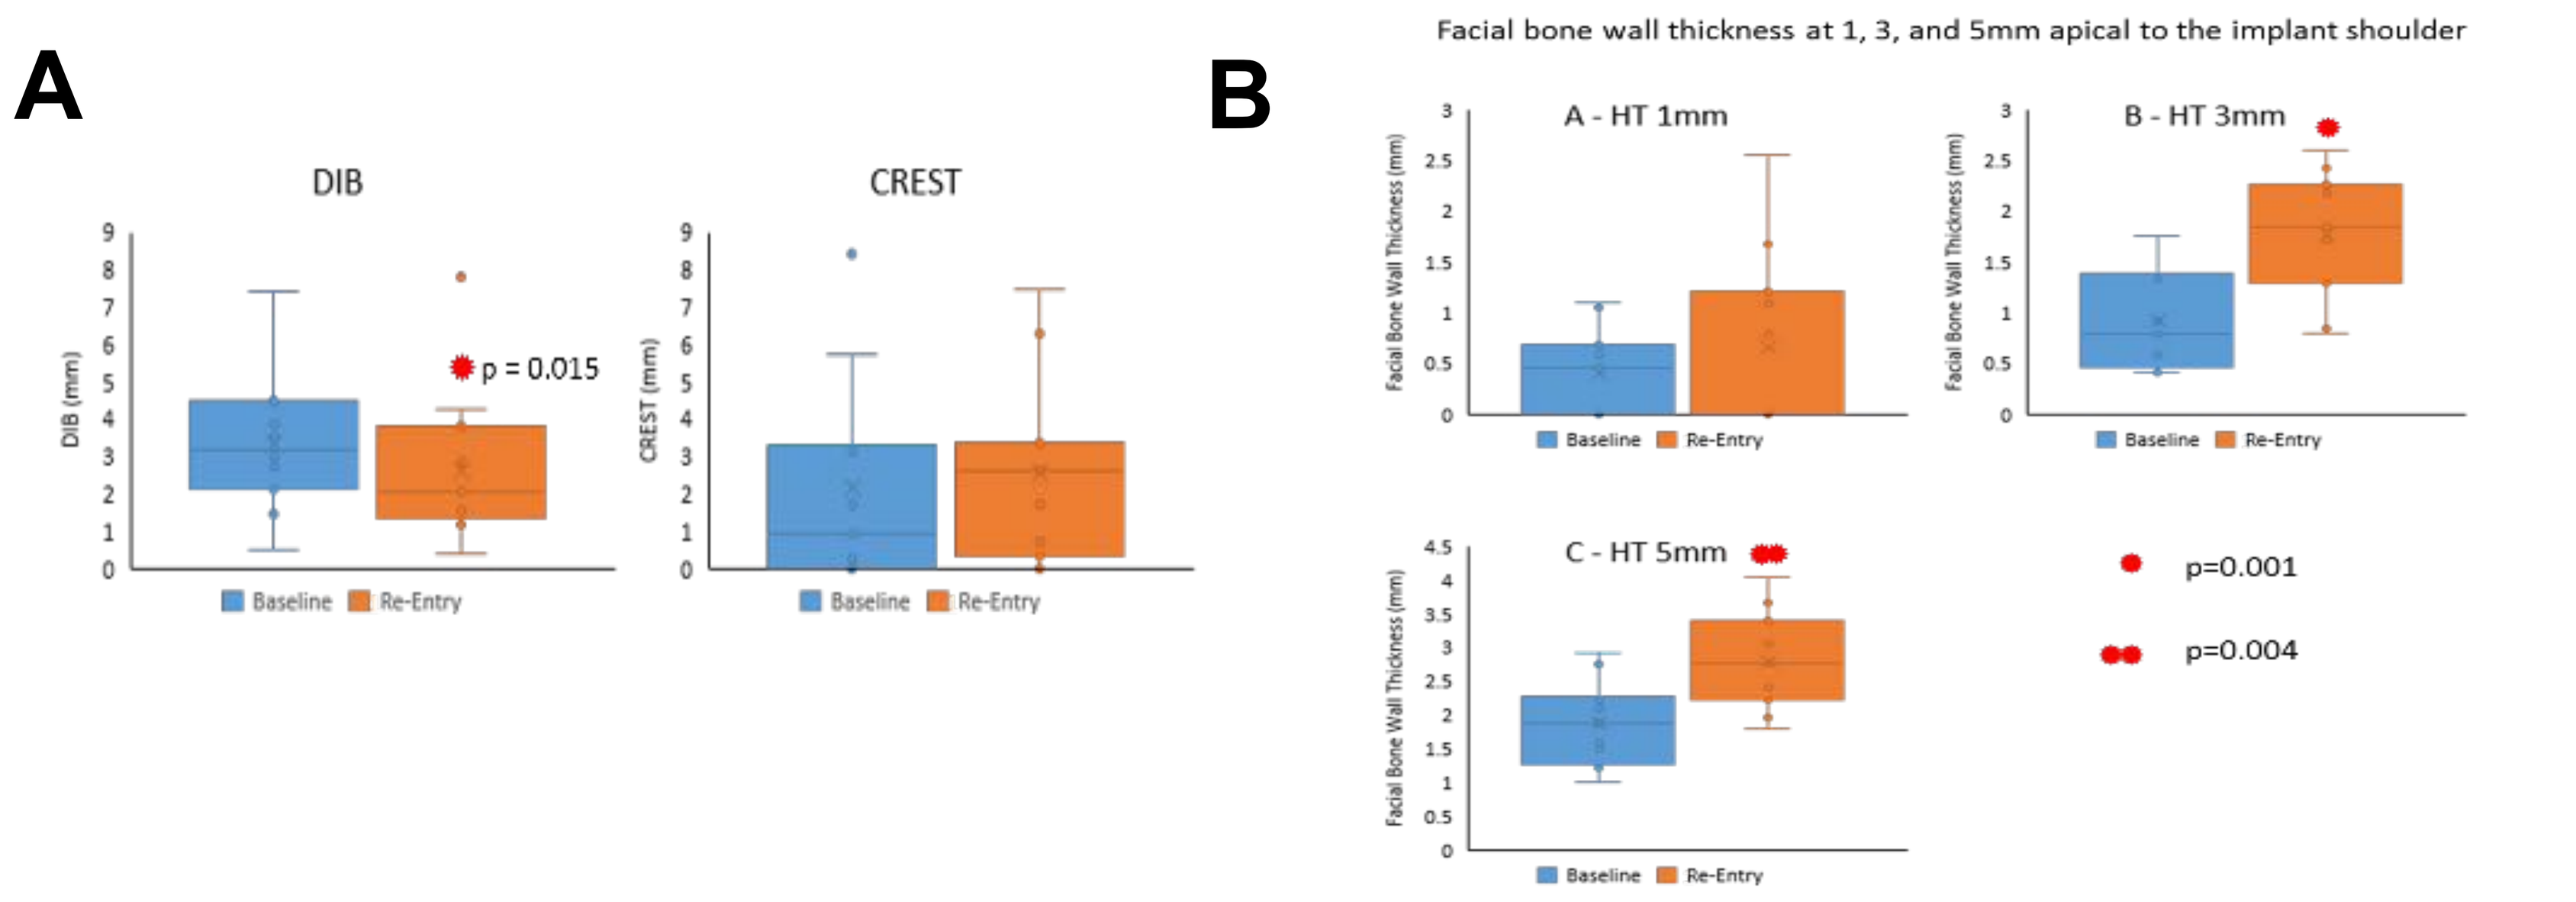
A
DIB
DIB (mm)
p = 0.015
CREST
CREST (mm)
Baseline
Re-Entry
B
Facial bone wall thickness at 1, 3, and 5mm apical to the implant shoulder
A - HT 1mm
B - HT 3mm
C - HT 5mm
Facial Bone Wall Thickness (mm)
p=0.001
p=0.004

Fig. 8. A. Vertical measurements of bone regeneration before and after GBR with CelGro™. DIB = distance from implant shoulder to first bone to implant contact. CREST = distance from top of the alveolar ridge to the first bone to implant contact. **B. Horizontal measurements (HT) of bone regeneration before and after GBR with CelGro™.** HT1/3/5 = horizontal thickness of the buccal alveolar crest (facial bone wall) measured at 1 (A), 3 (B), or 5mm (C) below the implant shoulder. HT values showed a statistically significant increase (*) at measurements taken 3mm (B) and 5mm (C) from the implant shoulder.

Conclusions

CelGro™ is an Australia-sourced porcine collagen membrane with excellent biocompatibility and handling characteristics. High quality bone was regenerated at all implant sites, resulting in increases in bone volume in both vertical and horizontal dimensions. The results of this study indicate that CelGro™ collagen membrane can be used in GBR treatment to preserve or restore bone volume required for successful functional and aesthetic outcomes in dental implant treatment.